IGV-001: A Biologic-Device Combination Product Elicits Potent Anti-GBM Responses as a Monotherapy and in Combination with Checkpoint Inhibition

Jenny Zilberberg,[1] Mark A. Exley[1]

[1]Imvax, Inc., Philadelphia, PA, USA

IMMU-30

INTRODUCTION

- Imvax is developing Goldspire®, a personalized immunotherapy that combines whole tumor–derived cells with an antisense oligonucleotide against insulin-like growth factor 1 receptor (IGF-1R; IMV-001) in biodiffusion chambers (BDCs)
- BDCs with a 0.1-µm pore are irradiated and implanted at abdominal sites for approximately 48 hours to deliver an antigenic payload and immunostimulatory factors that together train the immune system to attack tumor cells
- The lead product, IGV-001, was evaluated in newly diagnosed glioblastoma (ndGBM) patients in a phase 1b study (ClinicalTrials.gov, NCT02507583)
 - The median overall survival (OS) of highest exposure IGV-001-treated Stupp-eligible patients (n = 10) was 38.2 months compared with 16.2 months in standard-of-care-treated patients ($p = 0.044$)
- A Phase 2b study in ndGBM (ClinicalTrials.gov, NCT04485949) investigating a primary end point of progression-free survival (PFS) and key secondary end points of OS and safety, recently completed enrollment

METHODS

GBM MODEL

- An orthotopically implanted glioblastoma (GBM; luciferase-expressing GL261-luc2 mouse cell line) model was used to evaluate the efficacy of the murine version of IGV-001 (*m*IGV-001) alone or in combination with checkpoint inhibition using an anti-PD-1 monoclonal antibody (mAb)

PREPARATION OF *m*IGV-001

- To prepare *m*IGV-001, the following steps occurred:
 - GL261-luc2 cells were treated with IGF-1R antisense IMV-001
 - After treatment, the cells were resuspended in phosphate-buffered saline (PBS) and loaded into clinical-grade BDCs (0.1-µm pore size, polyvinylidene difluoride membrane) at a density of 1×10^6 cells per BDC
 - Last, the BDCs were irradiated with 5–6 gray

IMPLANTATION OF BDCS

- Mice received BDCs filled with PBS (control) or *m*IGV-001 (experimental) (fig. 1)
- BDCs were subcutaneously (s.c.) implanted in the flank for 2 days (48 hours), followed by explantation and wound closing

FIGURE 1

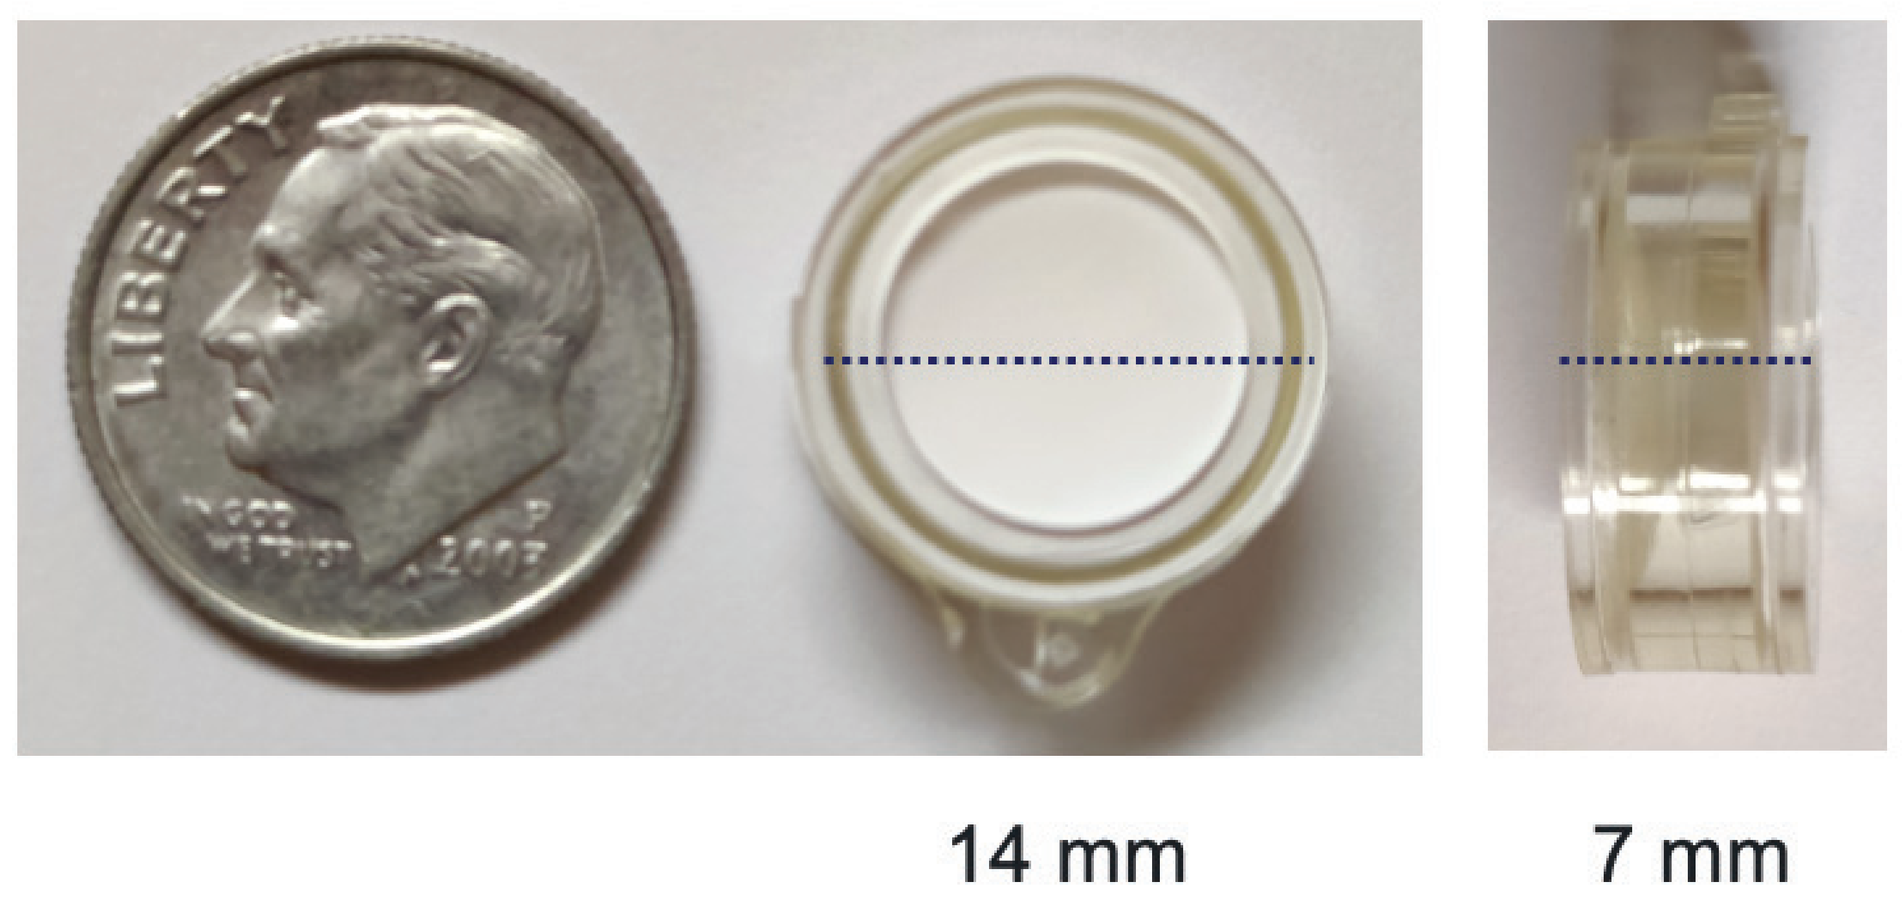
14 mm
7 mm

BDCs with a 0.1-µm pore and polyvinylidene difluoride membrane. Cells loaded at a density of 1×10^6 cells per BDC.
BDC, biodiffusion chamber.

TUMOR ESTABLISHMENT AND TREATMENT

- In the treatment setting, tumors were established for 7 days and then mice were randomized, by tumor size, prior to BDC implantation
- In preventative settings, on Day 26 after BDC explantation or mock surgery, mice received an orthotopic tumor challenge
- For both the treatment and preventative settings, mice also received either an anti-PD-1 antibody or an isotype control; the 4 treatment groups were:
 - Control (BDC plus PBS) + isotype mAb
 - *m*IGV-001 + isotype mAb
 - Control + anti-PD-1 mAb
 - *m*IGV-001 + anti-PD-1 mAb
- The anti-PD-1 mAb treatment was administered every 3 to 4 days for a total of 4 doses
- Following treatment, tumor progression and survival were monitored for an additional 100 days

RESULTS

TREATMENT SETTING

- In the treatment setting, mice receiving *m*IGV-001 in combination with an anti-PD-1 antibody experienced significantly longer progression-free survival after orthotopic tumor challenge than mice that received the control treatment (fig. 2)[1]

FIGURE 2

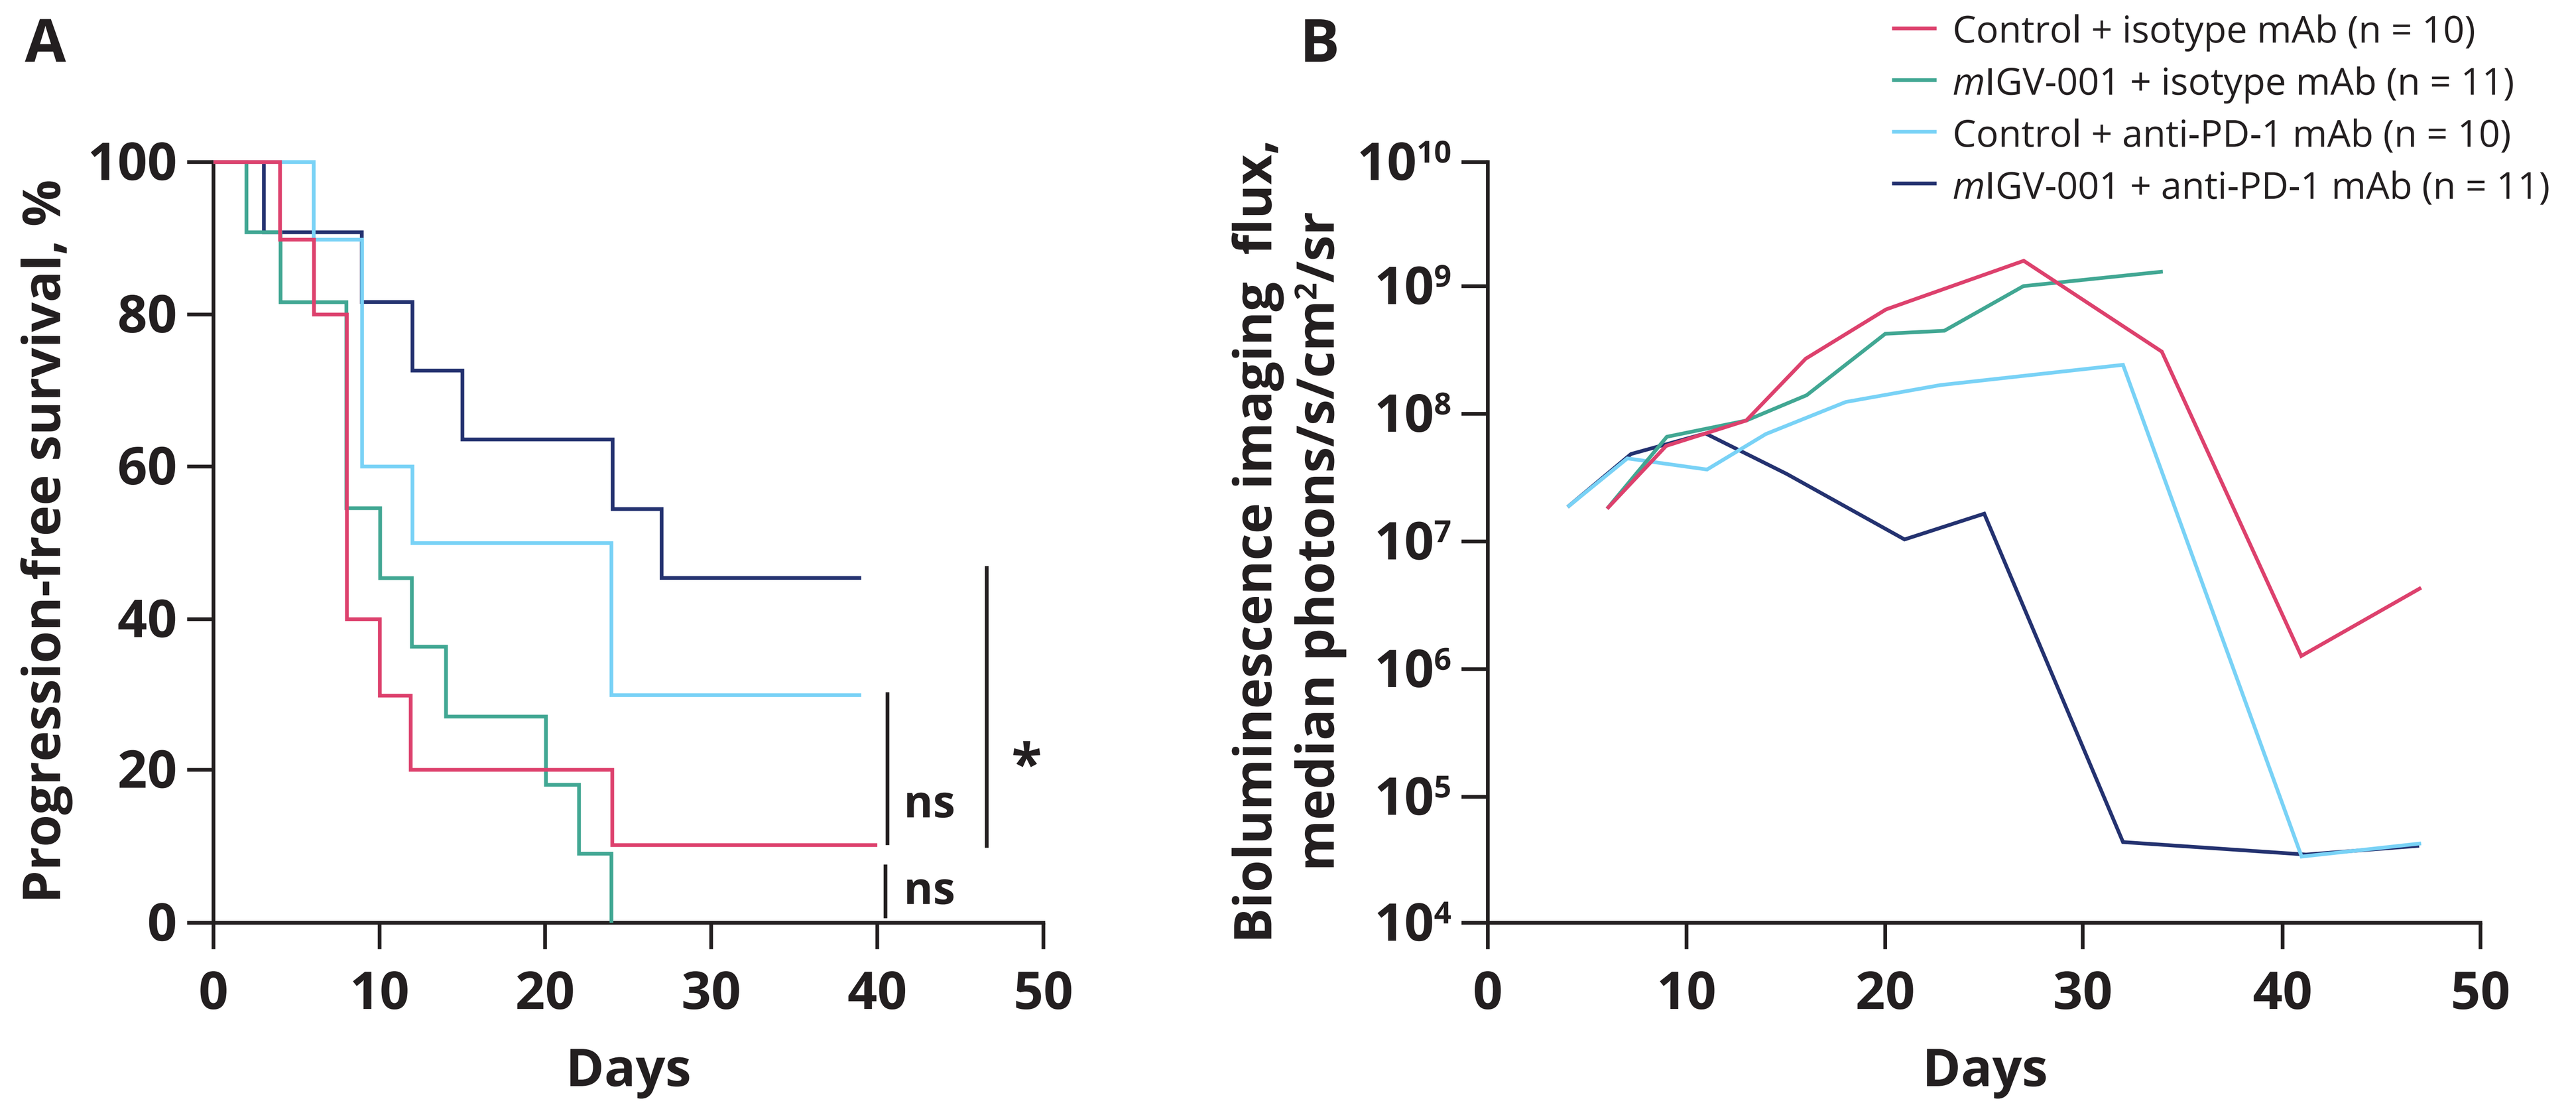
A
Progression-free survival, %
100
80
60
40
20
0
ns
ns
*
0
10
20
30
40
50
Days
B
Control + isotype mAb (n = 10)
mIGV-001 + isotype mAb (n = 11)
Control + anti-PD-1 mAb (n = 10)
mIGV-001 + anti-PD-1 mAb (n = 11)
Bioluminescence imaging flux, median photons/s/cm²/sr
10¹⁰
10⁹
10⁸
10⁷
10⁶
10⁵
10⁴
0
10
20
30
40
50
Days

Mice received intracranial tumor challenge and 7 days later were randomized into 4 different groups: (1) PBS-loaded BDC + isotype mAb; (2) *m*IGV-001 + isotype mAb; (3) PBS-loaded BDC + anti-PD-1 mAb; (4) *m*IGV001 + anti-PD-1 mAb. PBS-loaded BDC was used as control. *m*IGV-001 was manufactured with 1×10^6 GL261-luc2 cells/BDC. BDCs were implanted in the flank and left in place for 48 hours. (A) Kaplan-Meier progression-free survival curves. *Log-rank test, $p < 0.05$. (B) Median bioluminescence signal. Days = days post-intracranial tumor challenge. n = number of mice.

mAb, monoclonal antibody; *m*IGV-001, murine version of IGV-001; ns, not significant; PBS, phosphate-buffered saline; sr, steradian.

PREVENTATIVE SETTING

- Mice receiving preventative treatment with *m*IGV-001 in combination with anti-PD-1 antibody experienced significantly longer overall survival after orthotopic tumor challenge than mice receiving the control treatment (fig. 3)[2]

FIGURE 3

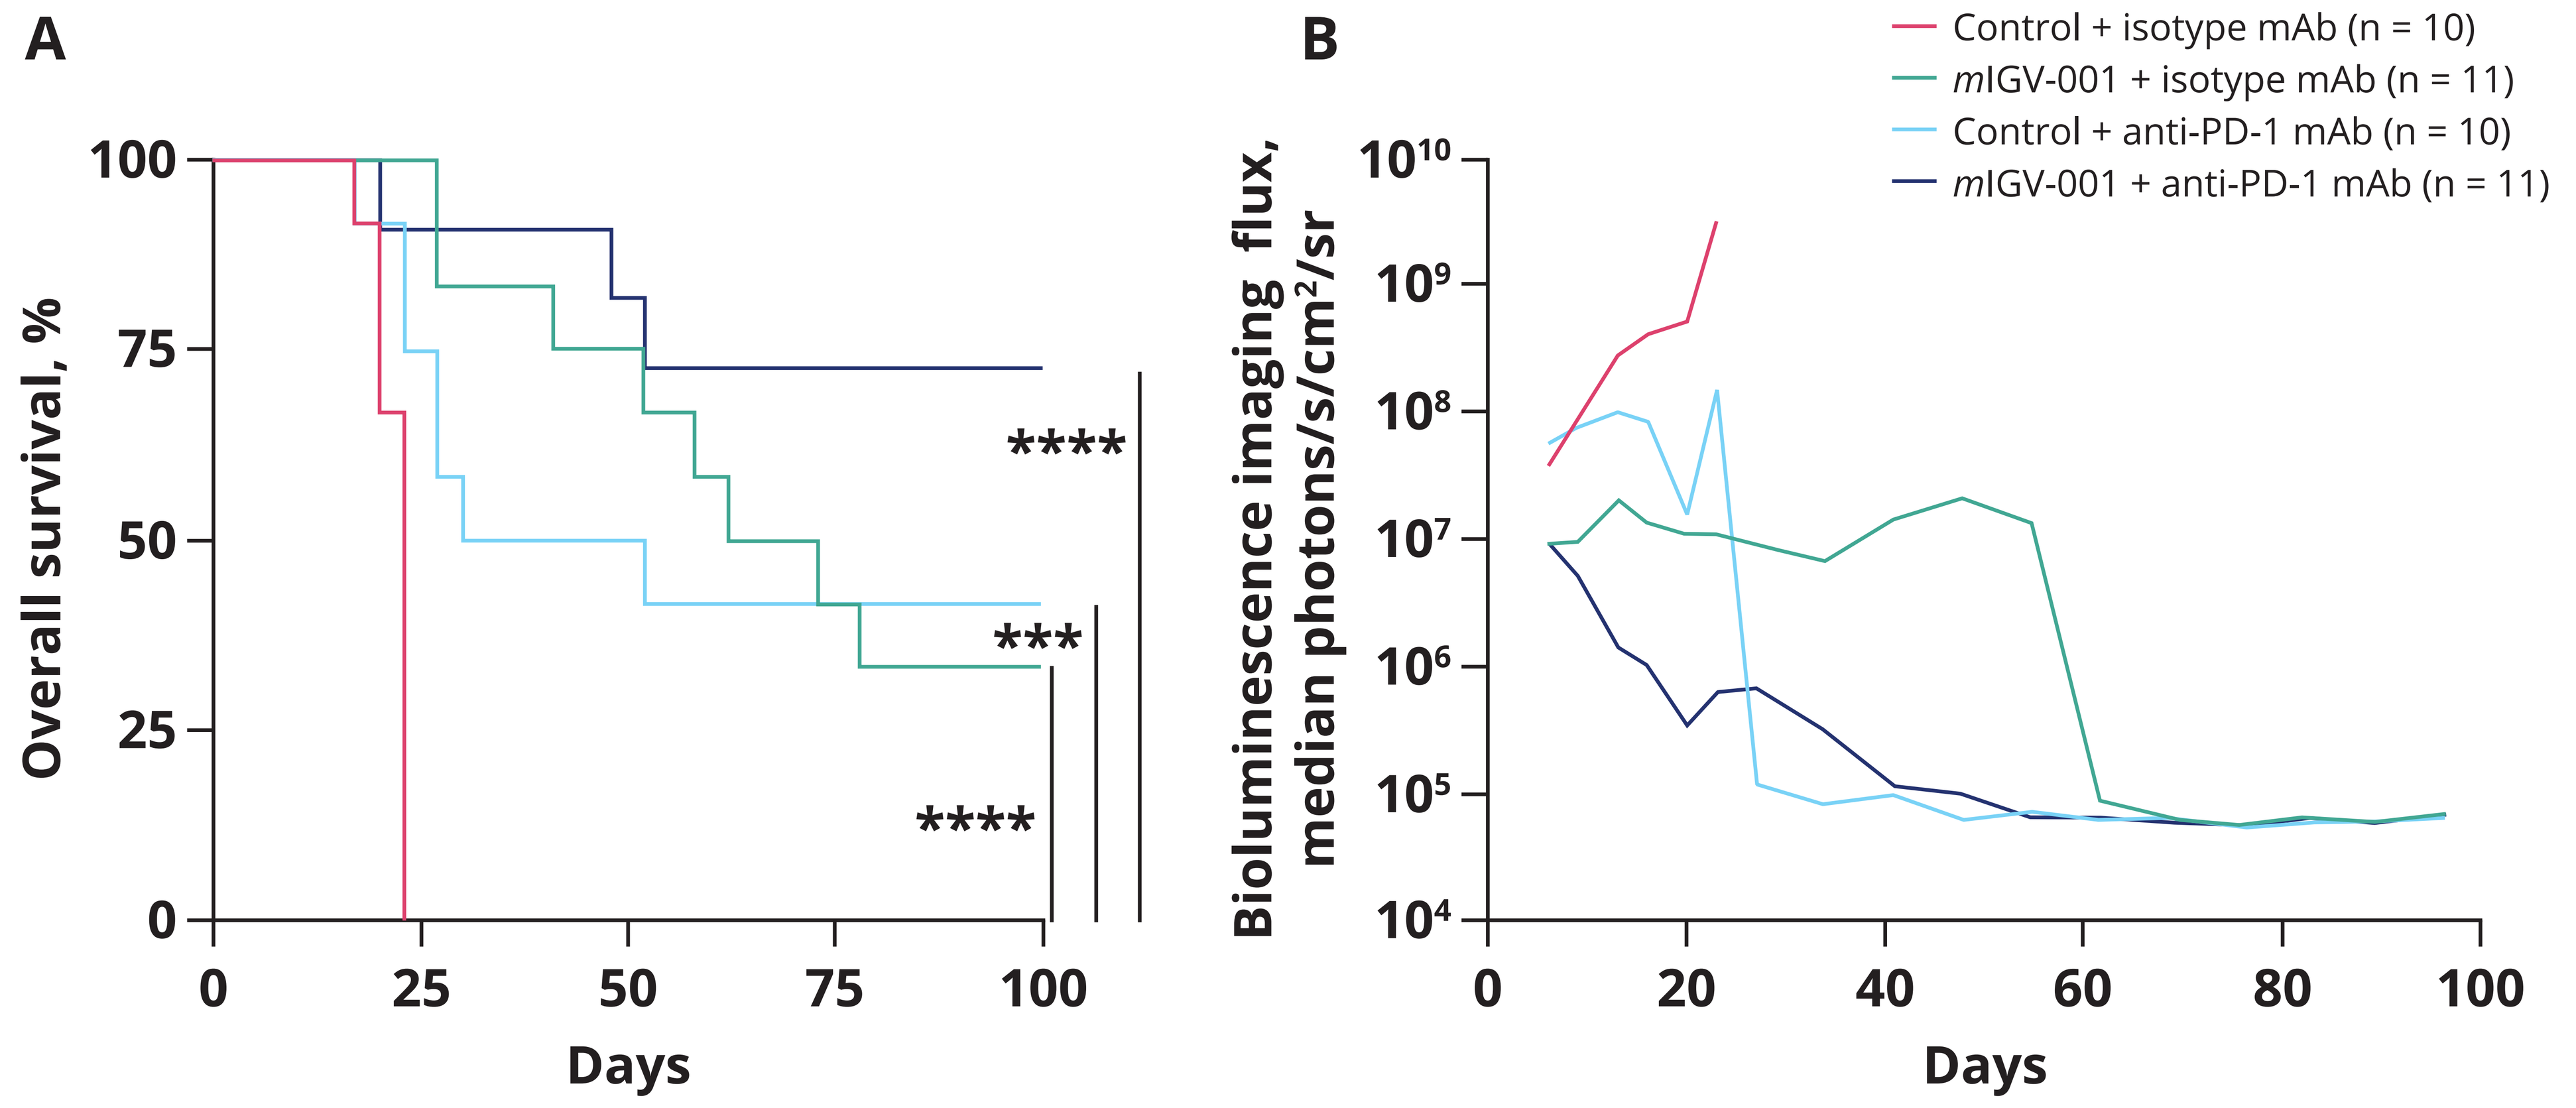
A
Overall survival, %
100
75
50
25
0

0
25
50
75
100
Days
B
Control + isotype mAb (n = 10)
mIGV-001 + isotype mAb (n = 11)
Control + anti-PD-1 mAb (n = 10)
mIGV-001 + anti-PD-1 mAb (n = 11)
Bioluminescence imaging flux, median photons/s/cm²/sr
10¹⁰
10⁹
10⁸
10⁷
10⁶
10⁵
10⁴
0
20
40
60
80
100
Days

BDCs were implanted in the flank and left in place for 48 hours. Orthotopic tumor challenges were performed 26 days after BDC explantation. Four different groups were evaluated: (1) PBS-loaded BDC + isotype mAb; (2) treatment (*m*IGV-001 or *m*IMC-001) + isotype mAb; (3) PBS-loaded BDC + anti-PD-1 mAb; (4) treatment (*m*IGV-001 or *m*IMC-001) + anti-PD-1 mAb. PBS-loaded BDC was used as control. (A) Kaplan-Meier survival curves of C57/BL6 albino female mice intracranially challenged with GL261-luc2 GBM cells. Log-rank, ****$p < 0.0001$, ***$p < 0.001$. (B) Median bioluminescence signal. Days = days post-intracranial tumor challenge. n = number of mice.

BDC, biodiffusion chamber; GBM, glioblastoma; mAb, monoclonal antibody; *m*IGV-001, murine version of IGV-001; ns, not significant; PBS, phosphate-buffered saline; sr, steradian.

CONCLUSIONS

- Mice receiving *m*IGV-001 + anti-PD-1 mAb after tumor establishment experienced greater progression-free survival and more durable control of GBM tumors versus monotherapy and placebo control groups
- Preventative treatment with *m*IGV-001 or anti-PD-1 mAb showed long-term benefit and was further enhanced with the combination *m*IGV-001 + anti-PD-1 mAb
- Further information can be accessed at: www.imvax.com

Acknowledgments
Medical writing and editorial support were provided by Citrus Scientific, a Citrus Health Group, Inc., company (Chicago, IL), in accordance with Good Publication Practices (GPP).

Disclosures
JZ and MAE: employee of Imvax Inc.

***Contact Information:**
Jenny Zilberberg, MS, PhD: J.Zilberberg@imvax.com

References
1. Cultrara C, et al. *J Immunother Cancer*. 2023;11(8):e006880.
2. Zilberberg C, et al. *Clin Immunol*. 2024;268:110373.

Poster presented at the 29th Annual Meeting for the Society for Neuro-Oncology (SNO). November 21–November 24, 2024, Houston, TX, USA

© Copyright 2024 IMVAX®, Inc. All rights reserved.